Plasma protein binding mediated DDI: A mathematical model is used to characterize drugs as potential displacers

Hinnerk Boriss[1], Oliver von Richter[2], Robert Hermann[3], Joachim Maus[4]

1) Sovicell GmbH, Deutscher Platz 5b, 04103 Leipzig, Germany, hbo@sovicell.com, 2) Merck Serono GmbH, 3) cr.appliance, 4) Meda Pharma GmbH

Introduction

A clinically relevant drug-drug interaction occurs when the effectiveness or toxicity of one medication is altered by the administration of another drug. Displacement of drugs from binding to plasma proteins can result in increased free concentrations of the displaced drug. This in turn increases the exposure and thus potential toxic effects at dosages that would otherwise be regarded as safe. The present work assesses specific binding properties of drugs which can lead to drug-drug interactions mediated by displacement form plasma protein binding.

Abstract

A differential equation model which describes the competitive binding of two ligands to two proteins each with a single binding site was constructed and analyzed. The model illustrated that only compounds with weak albumin binding ($K_D \geq 10^{-4}$) and strong AGP binding ($K_D \leq 10^{-6}$) can displace other compounds to a noticeable degree by at least doubling their free fraction. However, compounds that can displace other drugs from plasma binding can exert this potential only when the drug being potentially displaced is likewise bound only weakly to albumin and strongly to AGP. If not all of these criteria are met at the same time, displacement is impossible at pharmacologically relevant drug concentrations.
Flupirtine has been previously reported as a potent displacer of diazepam and warfarin. While new experimental data cannot confirm historical data, we show that the drug fulfills only one of the two essential criteria for plasma displacing drugs. Flupirtine binds only weakly to albumin, however, it exhibits no measureable affinity to AGP. Hence, the theoretical assessment of plasma displacement confirms recent experimental results.

Model

To characterize compound properties influencing plasma displacement a simulation model was constructed that describes the binding of two ligands with different properties to two proteins with one binding site each that can bind either ligand such that the ligands compete for binding at the binding sites on both proteins. The conceptual definition of the model is shown in Figure 1. The set of differential equations that describes this model is shown in Figure 2. Mathematica was used to analyze the model's properties.

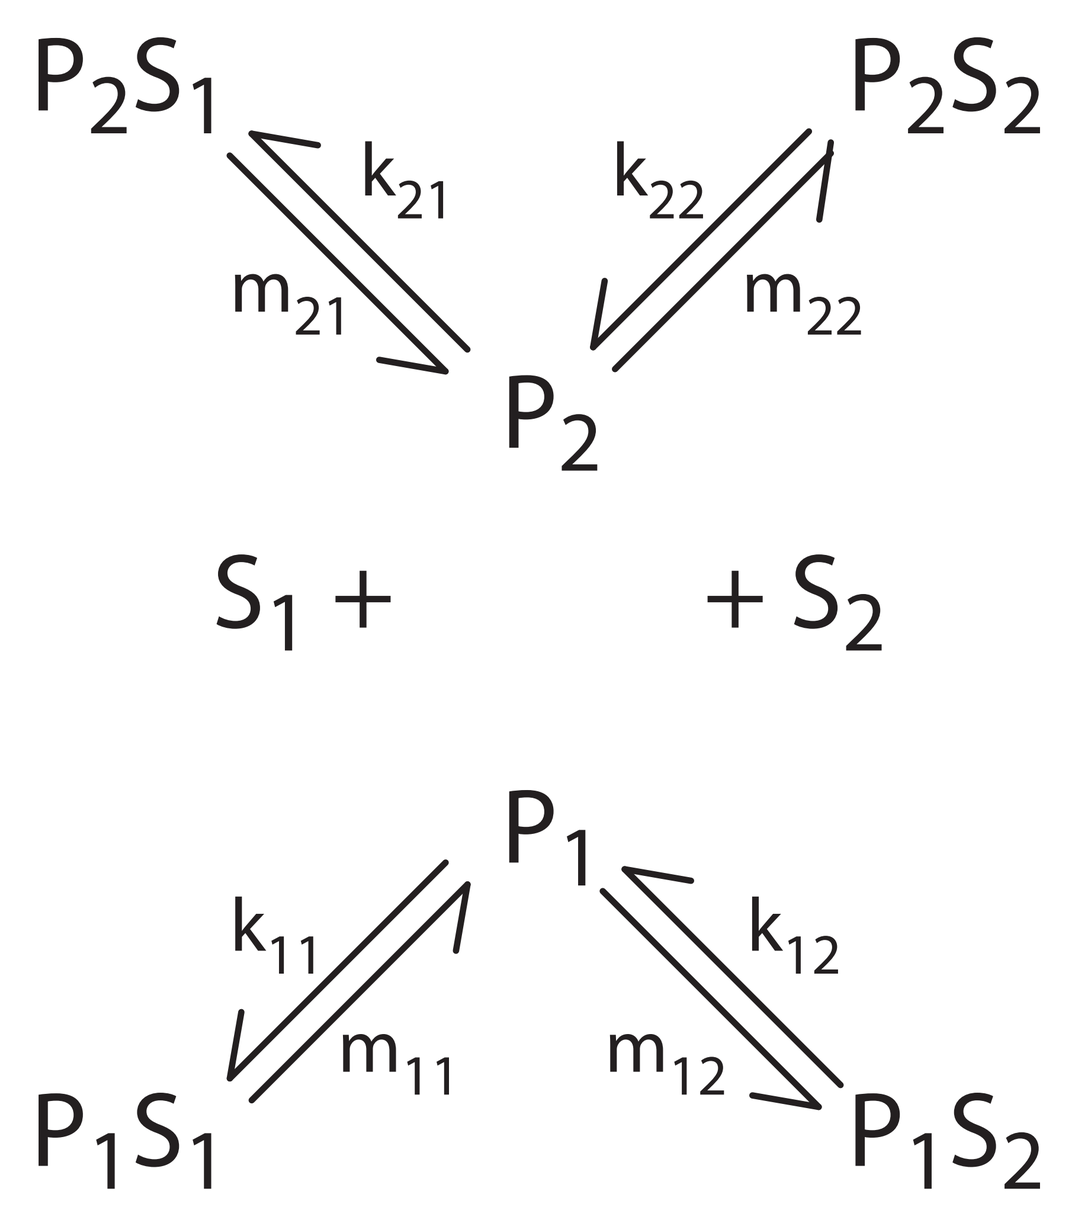

P2S1
P2S2
k21
m21
k22
m22
P2
S1 +
+ S2
P1
k11
m11
k12
m12
P1S1
P1S2

Fig. 1: Definition of the competitive binding model. Plasma proteins P_1 (=HSA) and P_2 (=AGP) each have a single binding site that can either bind ligand (drug) S_1 (flupirtine) or S_2 (test drug). The on-rates are denoted by k_{ij} while the off-rates are denoted by m_{ij}.

$$\frac{dS_1}{dt} = \mathrm{m}_{11}P_1S_1 - k_{11}\cdot S_1\cdot P_1 + m_{21}\cdot S_1 - k_{21}\cdot S_1\cdot P_2$$

$$\frac{dS_2}{dt} = \mathrm{m}_{12}P_1S_2 - k_{12}\cdot S_2\cdot P_1 + m_{22}\cdot S_1 - k_{22}\cdot S_2\cdot P_2$$

$$\frac{dP_1S_1}{dt} = k_{11}\cdot P_1\cdot S_1 - m_{11}\cdot P_1S_1$$

$$\frac{dP_1S_2}{dt} = k_{12}\cdot P_1\cdot S_2 - m_{12}\cdot P_1S_2$$

$$\frac{dP_2S_1}{dt} = k_{21}\cdot P_2\cdot S_1 - m_2\cdot P_2S_1$$

$$\frac{dP_2S_2}{dt} = k_{22}\cdot P_2\cdot S_2 - m_{22}\cdot P_2S_2$$

$$\frac{dP_1}{dt} = m_{11}\cdot P_1S_1 + m_{12}\cdot P_1S_2 - k_{11}\cdot P_1\cdot S_1 - k_{12}\cdot P_1\cdot S_2$$

$$\frac{dP_2}{dt} = m_{21}\cdot P_2S_1 + m_{22}\cdot P_2S_2 - k_{21}\cdot P_2\cdot S_1 - k_{22}\cdot P_2\cdot S_2$$

Fig. 2: Set of differential equations used to implement to model for simulation drug displacement. Ligand one is denoted by S_1 (denoted drug A in results section), ligand two is denoted by S_2 (denoted drug B in results section). HSA without ligand is denoted as P_1 and AGP without ligand is denoted P_2. HSA with ligand S_1 bound is denoted P_1S_1, AGP with ligand S_1 bound is denoted P_2S_1, HSA with ligand S_2 bound is denoted P_1S_2, AGP with ligand S_2 bound is denoted P_2S_2.

Experimental Results

Tab. 1: Summary of dialysis experiments with diazepam in human plasma and increasing concentrations of flupritine showing that the fraction unbound is independent of flupirtine.

compound name	incubation time [h]	flupirtine conc. [µM]	analytical method	fu [%]	recovery [%]
Diazepam	4	0.0	LC-MS/MS	8.1 ± 0.22	100.0
Diazepam	4	5.0	LC-MS/MS	7.8 ± 0.45	100.0
Diazepam	4	25.0	LC-MS/MS	8.5 ± 0.74	100.0

Tab. 2: Summary of dialysis experiments with warafrin in human plasma and increasing concentrations of flupritine showing that the fraction unbound is independent of flupirtine.

compound name	incubation time [h]	flupirtine conc. [µM]	analytical method	fu [%]	recovery [%]
Warfarin	4	0.0	LC-MS/MS	6.6 ± 0.74	100.0
Warfarin	4	5.0	LC-MS/MS	8.4 ± 1.72	100.0
Warfarin	4	25.0	LC-MS/MS	6.9 ± 0.71	100.0

Conclusion

- Plasma protein binding mediated drug-drug interation can only occur when special conditions are met (c.f. Table 3 and Figure 3):
 - Binding of potential displacer and displaced compound to alumin is weak
 - Binding of potential displacer and displaced compound to AGP, lipoproteins, or sex hormone binding protein is strong
- Flupitrine binds weakly to albumin, but not at all to AGP and thus cannot act as a displacer
- Experiemental data confirm that flupirtine doesn't displace diazeam (Table 1) or warfarin (Table 2) from plasma

Modelling Results

Tab. 3: Summary of displacement simulation outcomes from different combinations of binding characteristics of displaces and potentially displaced drugs (victim).

Case	HSA binding displacer/victim	AGP binding displacer/victim	Displacement
I	weak/weak	weak/weak	none
II	weak/strong	weak/weak	none
III	strong/weak	weak/weak	none
IV	strong/strong	weak/weak	none
VI	weak/weak	strong/strong	yes
VII	weak/weak	strong/weak	none
VIII	weak/weak	weak/strong	none

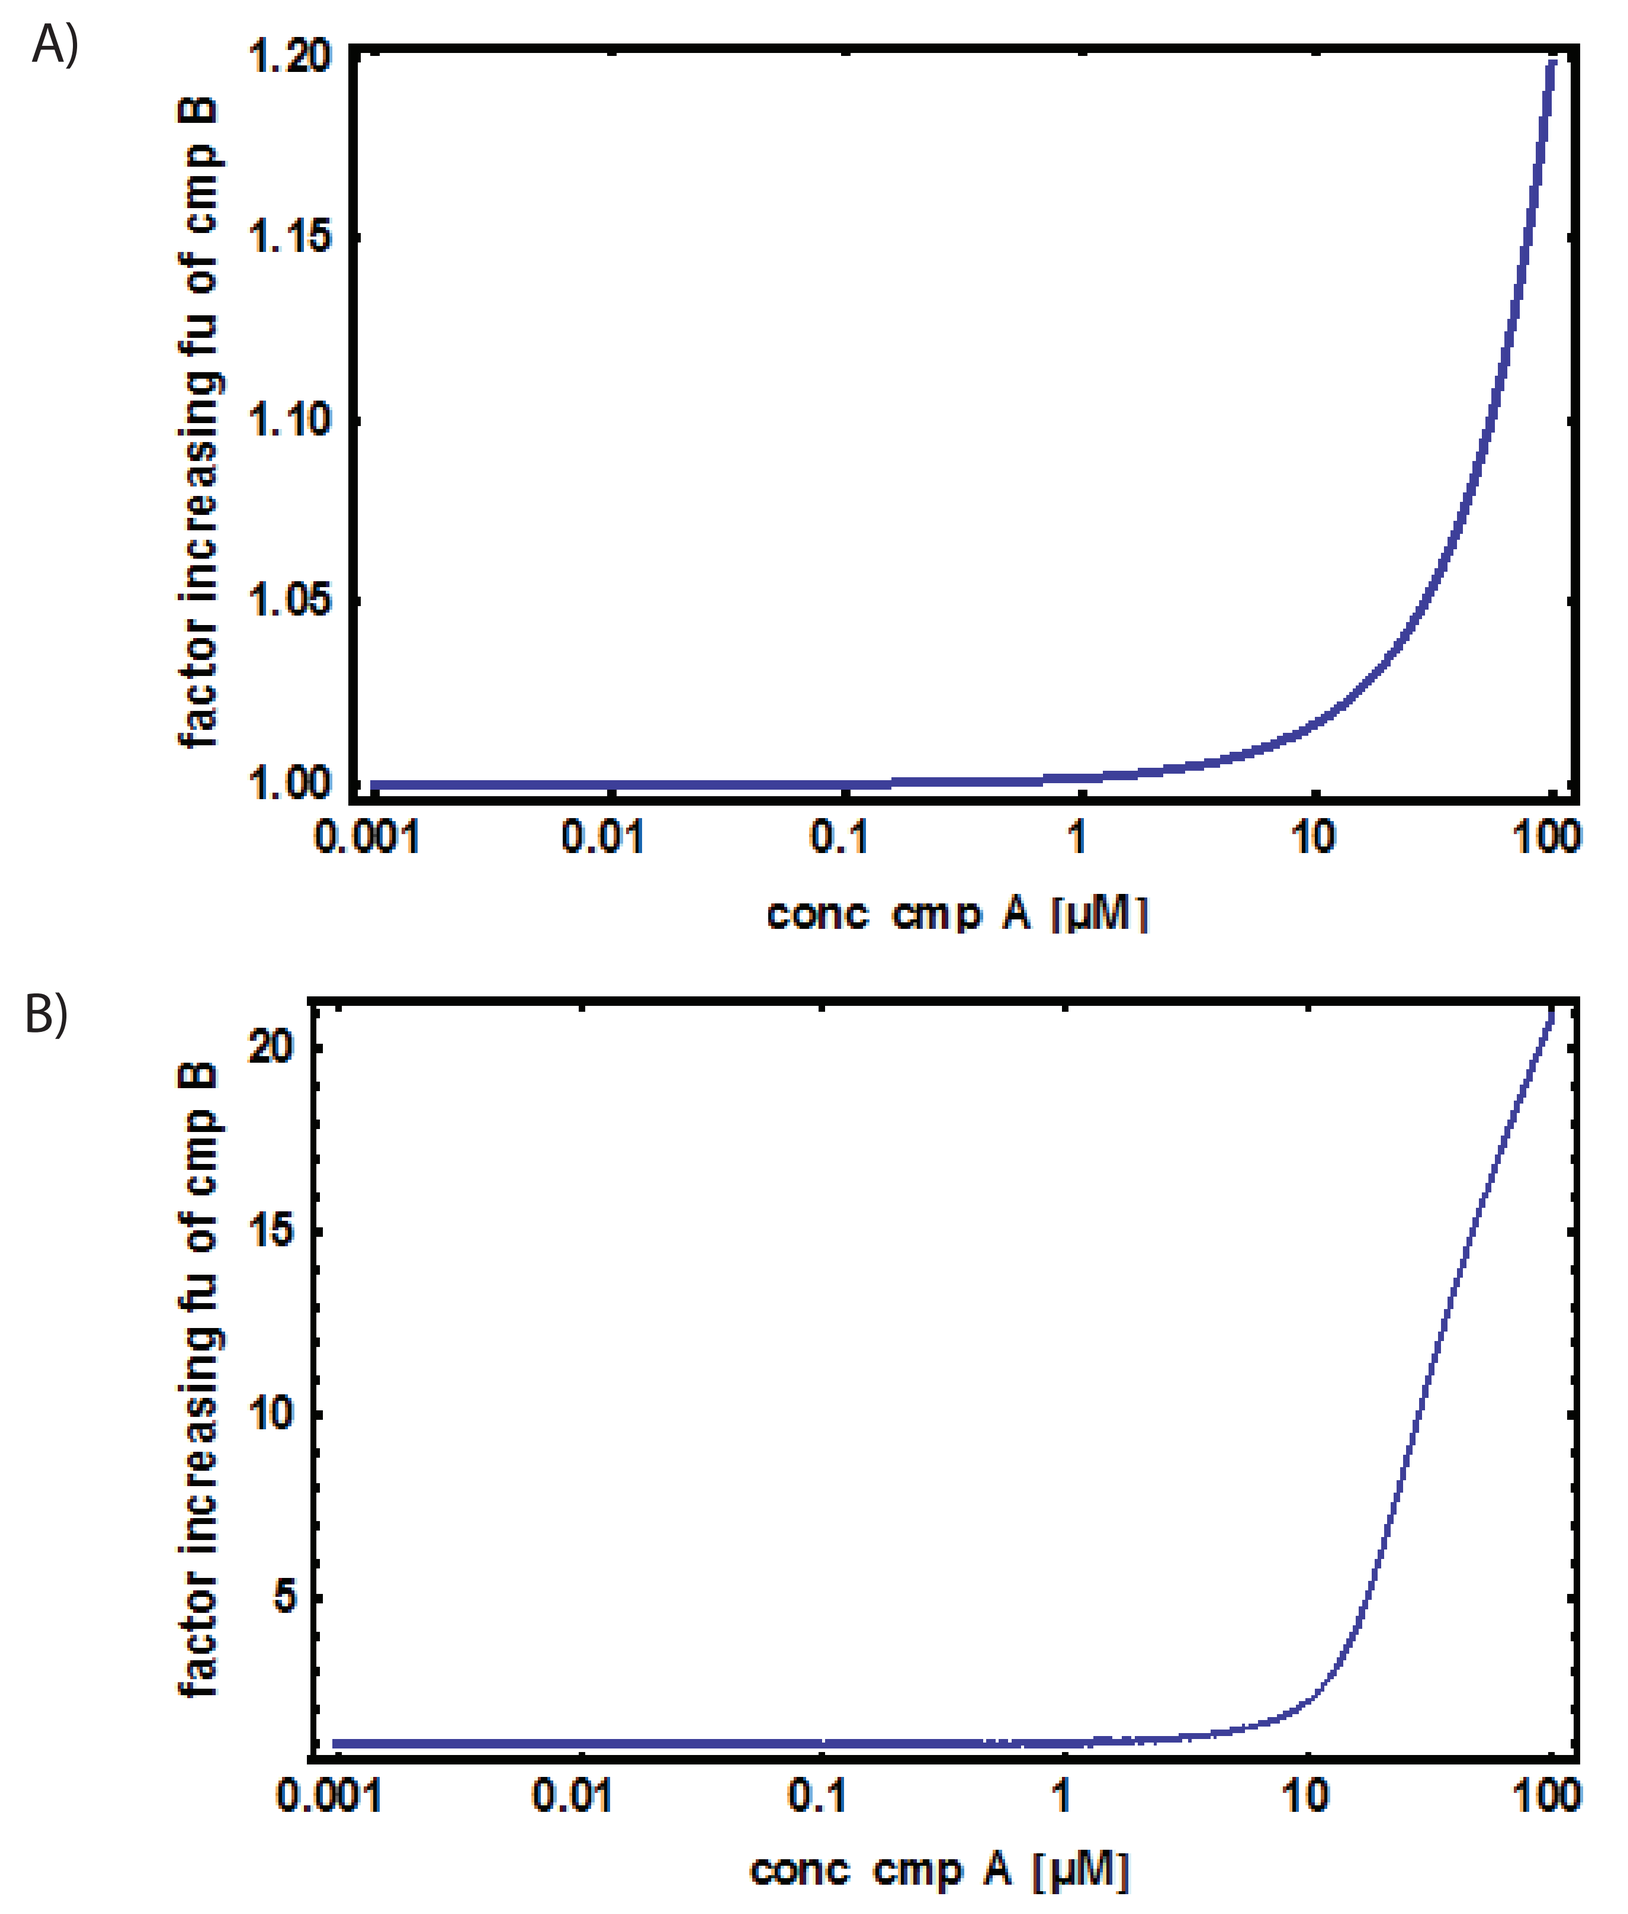

A)
factor increasing fu of cmp B
1.20
1.15
1.10
1.05
1.00
0.001
0.01
0.1
1
10
100
conc cmp A [µM]
B)
factor increasing fu of cmp B
20
15
10
5
0.001
0.01
0.1
1
10
100
conc cmp A [µM]

Fig. 3: Fold-change of the free fraction of the displaced drug in response to increasing the displacer concentration. A) HSA and AGP binding of both displacer and victim drug strong; B) HSA binding of displacer and victim drug weak and AGP binding of displacer and victim drug strong.

Sovicell
Deutscher Platz 5b 04103 Leipzig t. +49 341 520 44 0 f. +49 341 520 44 12 e. info@sovicell.com www.sovicell.com